Healing of Neuropathic Foot Ulcer using a Novel 'Wound Boot' (Kerraboot™)

Leigh R[1], Latif N[1], Hollingsworth S[2], Barker S[2], Hurel SJ[1]

Department of Diabetes[1] and Surgery[2], University College Hospitals, London, W1N 8AA.

Abstract: A 67-year-old man, with poorly controlled Type 2 diabetes, developed ulceration of both heels during a prolonged hospital admission for septicaemia. The left heel ulcer measured 8cm in maximum diameter and was approximately 1 cm deep (Figure 1). The right heel ulcer measured 3cm in maximum diameter and was approximately 0.3cm in depth. He had a peripheral sensory neuropathy. Doppler arterial ultrasound gave abnormally high ankle-brachial pressure indices, secondary to vessel calcification. Plain radiographs of the foot were suggestive of underlying osteomyelitis of the calcaneum. Magnetic resonance and nuclear imaging supported this diagnosis.

Despite bed rest, wound toileting and appropriate antimicrobials, the left heel ulcer continued to deteriorate with ulceration and infection spreading proximally to involve the posterior tibial compartment. To circumvent amputation, considered at the time to be the only suitable procedure, a novel '**wound boot**' was employed. The boot has been designed to provide a highly cost-effective, carer-friendly alternative to traditional dressings, whilst providing an optimal environment for wound healing. The boot was changed twice daily, often by the patient himself, and used in total for three weeks. Over this period, the infection resolved completely and the wound showed dramatic signs of healing (Figure 2).

INTRODUCTION

Diabetic neuropathic ulceration is extremely difficult to treat. Ischaemia and infection, affecting superficial tissues or the underlying bone, frequently complicate the condition. The management of these chronic ulcers is multidisciplinary; requiring scrupulous wound hygiene, careful choice and regular changing of dressings, systemic antibiotic therapy, and limb rest. Despite intensive management, healing time is frequently protracted and success rates are poor. The management of chronic diabetic foot ulcers makes considerable demands on nursing time, in hospitals and the community. [It has been estimated that over 25% of the district nurse's workload involves ulcer care.]

KERRABOOT

The **Kerraboot** 'wound boot' (Ark Therapeutics) is a tri-laminar, foot-shaped plastic boot, which fastens securely around the leg to enclose the area of ulceration. The key elements of the **Kerraboot** are:

- Warm, moist, protected environment – to promote wound granulation and healing,
- Super-absorbent padding – to remove excess moisture and wound exudate,
- Integral; charcoal filter and carbon impregnated material – to eliminate odour.
- Textured base – to prevent slipping during patient mobilisation.

CASE REPORT

- Patient – male, 67 years.

MEDICAL HISTORY

- 1983 Type 2 diabetes – glibenclamide (10 mg o.d.)
- Poor glycaemic control – glycosylated haemoglobin 12.5% (normal 4-6%)
- Dyslipidaemia – bezafibrate
- Regular alcohol consumption – 6 units daily
- 1987 progressive neuropathy – initially left foot, then both feet
- Other diabetic complications – proliferative retinopathy and nephropathy
- 1996 CVA – persistent left hemi-paresis
- June 1999 – Gram-negative septicaemia (secondary to UTI) - prolonged hospitalisation
- **Development of pressure sores to heels** – discharged, ulcer care by District Nurse and Podiatry services
- **October 1999 significant deterioration of both ulcers – re-admitted to hospital**

INVESTIGATIONS

Both ulcers produced a brown, foul-smelling exudate. The right ulcer was 3cm in diameter and 0.3cm deep; it showed no sign of cellulitis. The left ulcer measured approximately 8cm in diameter and was 1cm deep. The surrounding tissue was very macerated and there was local cellulitis.

Doppler arterial ultrasound – high ankle-brachial pressure indices (e.g. Dorsalis pedis: L >300mmHg, R 240mmHg), probably secondary to vessel calcification.

Microbiology – initial wound cultures grew *Pseudomonas aeruginosa* and *Staphylococcus aureus*.

Radiography, MR and nuclear imaging – osteomyelitis of calcaneum.

Biochemistry – C-reactive protein (CRP) 94, erythrocyte sedimentation rate (ESR) 80, white cell count (WCC) 8.3 x 10^9/L.

INITIAL MANAGEMENT OF PATIENT'S HEEL ULCERS

Surgical debridement
IV antibiotics (ceftazidime, flucloxacillin, ciprofloxacin, metronidazole)
Wound care (dry dressings changed every 2 days)
Strict bed rest

At 4 Weeks
Right heel ulcer healed
Left heel ulcer margin contracting
CRP fallen to 70

At 5 Weeks
Deterioration left heel ulcer
MRSA detected (teicoplanin given)
Pyrexia, CRP 213

Amputation discussed
Patient consented to try Kerraboot

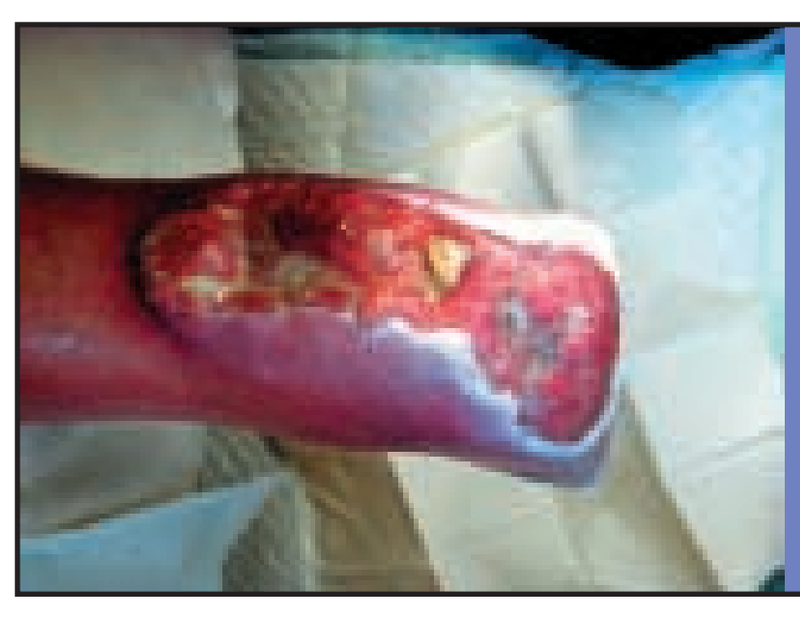

Kerraboot protocol introduced

MANAGEMENT OF LEFT HEEL ULCER WITH KERRABOOT

The **Kerraboot** was applied twice daily for 24 days.
The patient was easily able to change the boot, after simple instruction.
He was able to move freely.

After 14 days – notable ulcer healing
CRP 39.4
WCC 7.5 x 10^9/L.

By 21 days
ulcer margins contract and granulation at base of ulcer; CRP 12.7
WCC 7.3 x10^9/L.

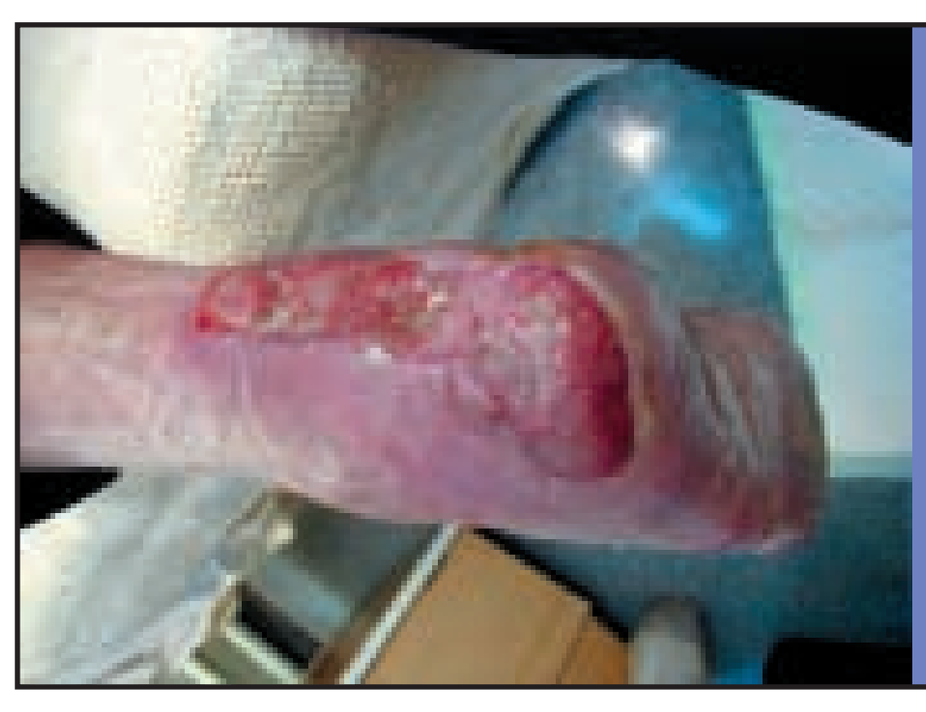

Patient discharged
– with weekly review at diabetes podiatry clinic.
Antibiotics – rifampicin and fusidic acid.

POTENTIAL BENEFITS OF THE KERRABOOT FOR DIABETIC FOOT ULCERS

- Warm, moist, protected environment to promote wound healing
- Super-absorbent material to absorb exudate and excessive moisture
- Transparent material to allow easy monitoring of the ulcer
- Efficient odour control to improve patient and ward acceptability
- Ease of use – patient or carer can change Kerraboot with minimum of tuition
- No other dressings required
- Potential cost benefits from reduced hospitalisation and nursing workload
- Aids mobilisation of the patient
- Opportunity to increase patient involvement in wound management.

CONCLUSION

This intervention appeared to promote healing of a chronic, infected, necrotic, neuropathic heel ulcer in a diabetic patient, and circumvented the need for amputation.

The evidence from this case suggests that the Kerraboot produces an environment conducive to wound healing. A study is currently underway to confirm these results.

Ark Therapeutics

Supported by an educational grant from Art Therapeutics
6 Warren Mews, London W1T 6AR Telephone. 020 7388 7722